A Placebo-Controlled Phase I Study in Healthy Subjects Assessing NVG-291 which Targets CNS Receptor Protein Tyrosine Phosphatase Sigma (PTPσ)

Marc DePaul, Betty Lawrence, Nana Collett, Judy Toews, Matvey Lukashev, Daniel D. Mikol

Background

- Chondroitin sulfate proteoglycans (CSPGs) accumulate at sites of CNS damage and inhibit CNS repair
- A CSPG receptor termed protein tyrosine phosphatase sigma (PTPσ) has been implicated in mediating inhibitory effects of CSPGs
- NVG-291 is a subcutaneously (SC) administered peptide mimetic of the wedge domain of PTPσ, the rodent version of which has been shown to promote neural repair in various animal models of nervous system damage
- Across these models, NVG-291 treatment has resulted in improved functional outcomes when administered for 1-7 weeks as a daily subcutaneous injection, with histologic evidence of enhanced axonal regeneration, remyelination and neuroplasticity
- **Hypothesis**: Enhanced plasticity resulting from NVG-291 treatment will be reflected by changes in functional brain imaging measures in Alzheimer's Disease (AD)

Methods

- Phase 1 study design: Part 1 Single Ascending Dose (SAD); Part 2 Multiple Ascending Dose (MAD) (**Figure 1, Table 1**)

Figure 1. Dosing schedule

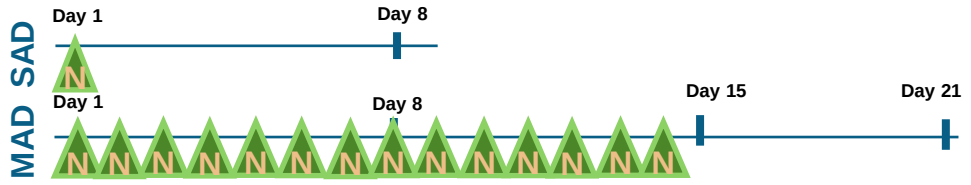
SAD
MAD
Day 1
Day 8
Day 1
Day 8
Day 15
Day 21

Table 1. Dose level cohorts in SAD and MAD

Dose level	Dose (mg/kg) 20 mg/mL solution	SAD Cohorts Single SC inj.	MAD Cohorts Daily SC inj. (14 days)
1	**0.032**	SAD1 1 NVG-291 1 Placebo	
2	**0.096**	SAD2 4 NVG-291 2 Placebo	
3	**0.192**	SAD3 4 NVG-291 2 Placebo	
4	**0.384**	SAD4 4 NVG-291 2 Placebo	MAD1 4 NVG-291 2 Placebo
5	**0.576**	SAD5 4NVG-291 2 Placebo	MAD2 4 NVG-291 2 Placebo
6	**0.864**	SAD6 7 NVG-291 4 Placebo	MAD3 4 NVG-291 2 Placebo

- The doses tested exceed the human equivalent doses that showed efficacy in animal models: 11 – 500 µg/day in rats (Human equivalent range **0.007 – 0.32 mg/kg**)

Results

- NVG-291 has been safe and well-tolerated through 6 SAD cohorts (unblinded) and 2 MAD cohorts (blinded analysis)
- Currently enrolling MAD cohort 3
- Most common adverse event (AE): injection site-related (**Table 2**; all mild; more common in pooled NVG-291 group); otherwise, no AE clearly increased in the NVG-291 group vs. placebo
- No effects on vital signs, electrocardiograms (ECG)
- A single serious AE has occurred in MAD cohort 3, not yet unblinded: mild increase of troponin level resulting in overnight hospitalization, without ECG changes
- Maximum concentration achieved < 1 hour; longer half-life than observed in nonclinical animal species (**Figure 2**)

Table 2. Adverse Events in SAD cohorts

	Pooled NVG-291	**Pooled Placebo**
Subjects with ≥ 1 TEAE	N=24 24 (100%)	N=13 7/13 (54%)
General Disorders and administration conditions	24 (100%)	7 (54%)
Injection site erythema	24 (100%)	6 (46%)
Injection site pain	16 (67%)	1 (8%)
Injection site pruritus	6 (25%)	0
Injection site edema	2 (8%)	0
Injection site paresthesia	2 (8%)	0
Injection site swelling	2 (8%)	0
Injection site bruising	1 (4%)	1 (8%)
Injection site mass	1 (4%)	0
Fatigue`	0	1 (8%)
Nervous system Disorders	7 (29%)	4 (31%)
Headache	6 (25%)	2 (15%)
Dizziness	2 (8%)	1 (8%)
Head discomfort	1 (4%)	0
Presyncope	1 (4%)	0
Lethargy	0	1 (8%)
Other		
Rash macular	1 (4%)	0
Skin odor abnormal	1 (4%)	0
Diarrhea	1 (4%)	0
Upper respiratory tract infection	1 (4%)	0
Nasal congestion	1 (4%)	0
Rhinorrhea	0	1 (8%)
Cough	0	1 (8%)
Muscle spasms	1 (4%)	0
Oropharyngeal discomfort	0	1 (8%)
Dysmenorrhoea	0	1 (8%)

Table does not include singular AEs related to blood draws or IV catheters

Figure 2. Average plasma concentrations of NVG-291 in SAD / MAD cohorts

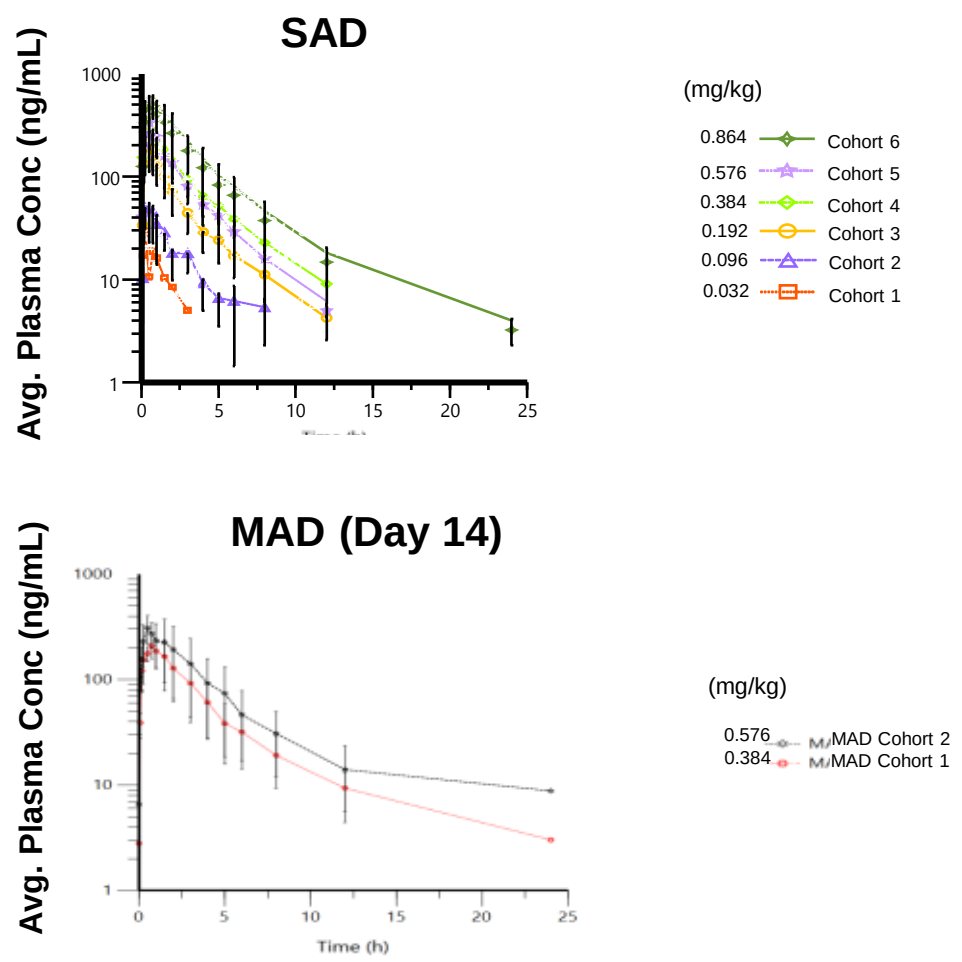
SAD
Avg. Plasma Conc (ng/mL)
1000
100
10
1
0
5
10
15
20
25
(mg/kg)
0.864 Cohort 6
0.576 Cohort 5
0.384 Cohort 4
0.192 Cohort 3
0.096 Cohort 2
0.032 Cohort 1
MAD (Day 14)
Avg. Plasma Conc (ng/mL)
Time (h)
(mg/kg)
0.576 MAD Cohort 2
0.384 MAD Cohort 1

- Lower limit of quantification 2 ng/mL
- Error bars show standard deviations

Discussion

- Results from the Phase 1 study to date are supportive of proceeding to a 24-week Phase 1b/2a placebo-controlled clinical trial in AD (**Figure 3**)
- Phase 1b/2a study objectives
 - **Primary objective**
 - Safety, tolerability, and pharmacokinetics of NVG-291
 - **Secondary objectives**
 - Change in cerebral glucose metabolism:
 1) Statistical region of interest (sROI) analysis
 2) Voxel-based subtraction analysis
 - Change in cognition (ADAS-Cog 13)
 - Global impression of change (CIBIC-plus)
 - **Exploratory objectives** will include assessment of additional cognitive measures, advanced structural and functional (including resting state connectivity using MRI-BOLD) imaging metrics, and plasma/CSF biomarkers

Figure 3. Phase 1b/2a study design

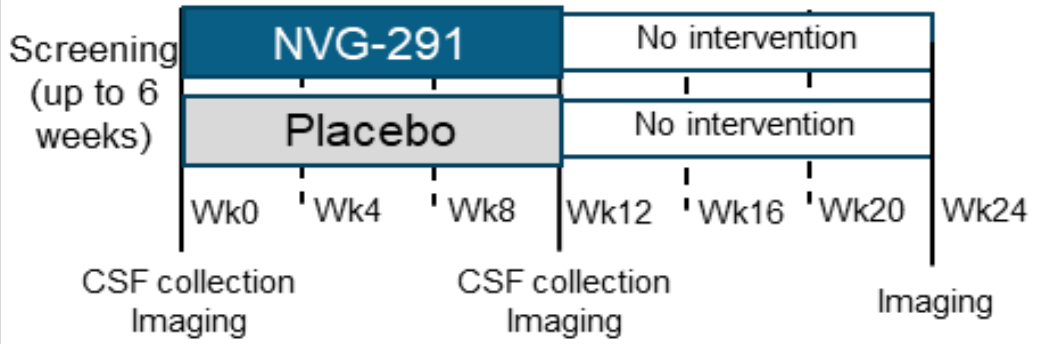
Screening (up to 6 weeks)
NVG-291
No intervention
Placebo
No intervention
Wk0
Wk4
Wk8
Wk12
Wk16
Wk20
Wk24
CSF collection Imaging
CSF collection Imaging
Imaging

- Weeks 1-12: Daily SC injection of NVG-291 (fixed dose) or placebo
- Weeks 13-24: No intervention; Rationale: Assess durability of NVG-291 effect and reduce placebo effect (week 24 assessments)
- **Key eligibility criteria**:
 - Mild cognitive impairment (MCI) or mild dementia due to AD
 - Age 55-85
 - Mini-mental state exam (MMSE) score 22 - 28
 - Abnormal paragraph recall to identify a well-defined amnestic population
 - Plasma phospho-tau-181 positive (step 1, enrichment)
 - CSF beta-amyloid positive (step 2, confirmation of AD)
 - Hypometabolism in sROI on 18F-fluorodeoxyglucose (^{18}FDG)-positron emission tomography (step 3)
- **Sample size**: The sample size (N~80) in this Phase 1b/2a study is deemed sufficient to assess the safety of NVG-291 in an AD population
 - There are no pre-determined expectations for efficacy based on functional imaging, clinical measures, structural imaging or fluid biomarkers
- **Number of sites**: 15-20

Conclusion

Results of the Phase 1 study will establish the safety, tolerability, and pharmacokinetics of NVG-291 to support advancement to a Phase 1b/2a clinical trial in patients with MCI or mild dementia due to AD in 2022